Zebrafish larvae as toxicity model for drug development using imaging mass spectrometry

Junhai Yang, James Sawicki, Nari Talaty, Steven Cassar, Lauren Rendino, Sabyasachi Biswas, Wayne Buck, David Wagner

Introduction

Introduction
Organ specific distribution can impact efficacy, toxicity, and drug level kinetics. Using the zebrafish model to predict tissue distribution of compounds could allow early assessment of this characteristic and allow comparisons between compounds early in compound selection to optimize for ideal distribution at lower cost and a quicker turnover. Mass spectrometry imaging provides a way to determine drug level through virtual dissection of these microscopic specimens.
This poster presents the method development of zebrafish larvae embedding/ sectioning, and imaging result of toxicity model compounds dosed zebrafish larvae.

Method

Zebrafish of 7 days-post-fertilization were dosed separately with four compounds (clozapine, diltiazem, buspirone and loperamide), and placed on the fish shaped surface of solid gelatin block (20%) made from 3D printed mold, the site of larvae was covered by liquid gelatin (20%). Sections of 10 µm thickness obtained using a HM550 Cryostat at -16°C were coated with DHB on an HTX TM sprayer or sublimation. MALDI Imaging was carried out on a Bruker 7T Solarix FT-ICR or a timsTOF Flex MALDI 2 mass spectrometer. Following imaging acquisition, H&E optical images of the same section was obtained on a Meiji microscope MT4300LV or a Motic slide scanner EasyscanPro1. In parallel, DESI mass spectrometry imaging was applied to the whole fish to obtain the DESI ion images.

MALDI imaging of zebrafish larvae workflow

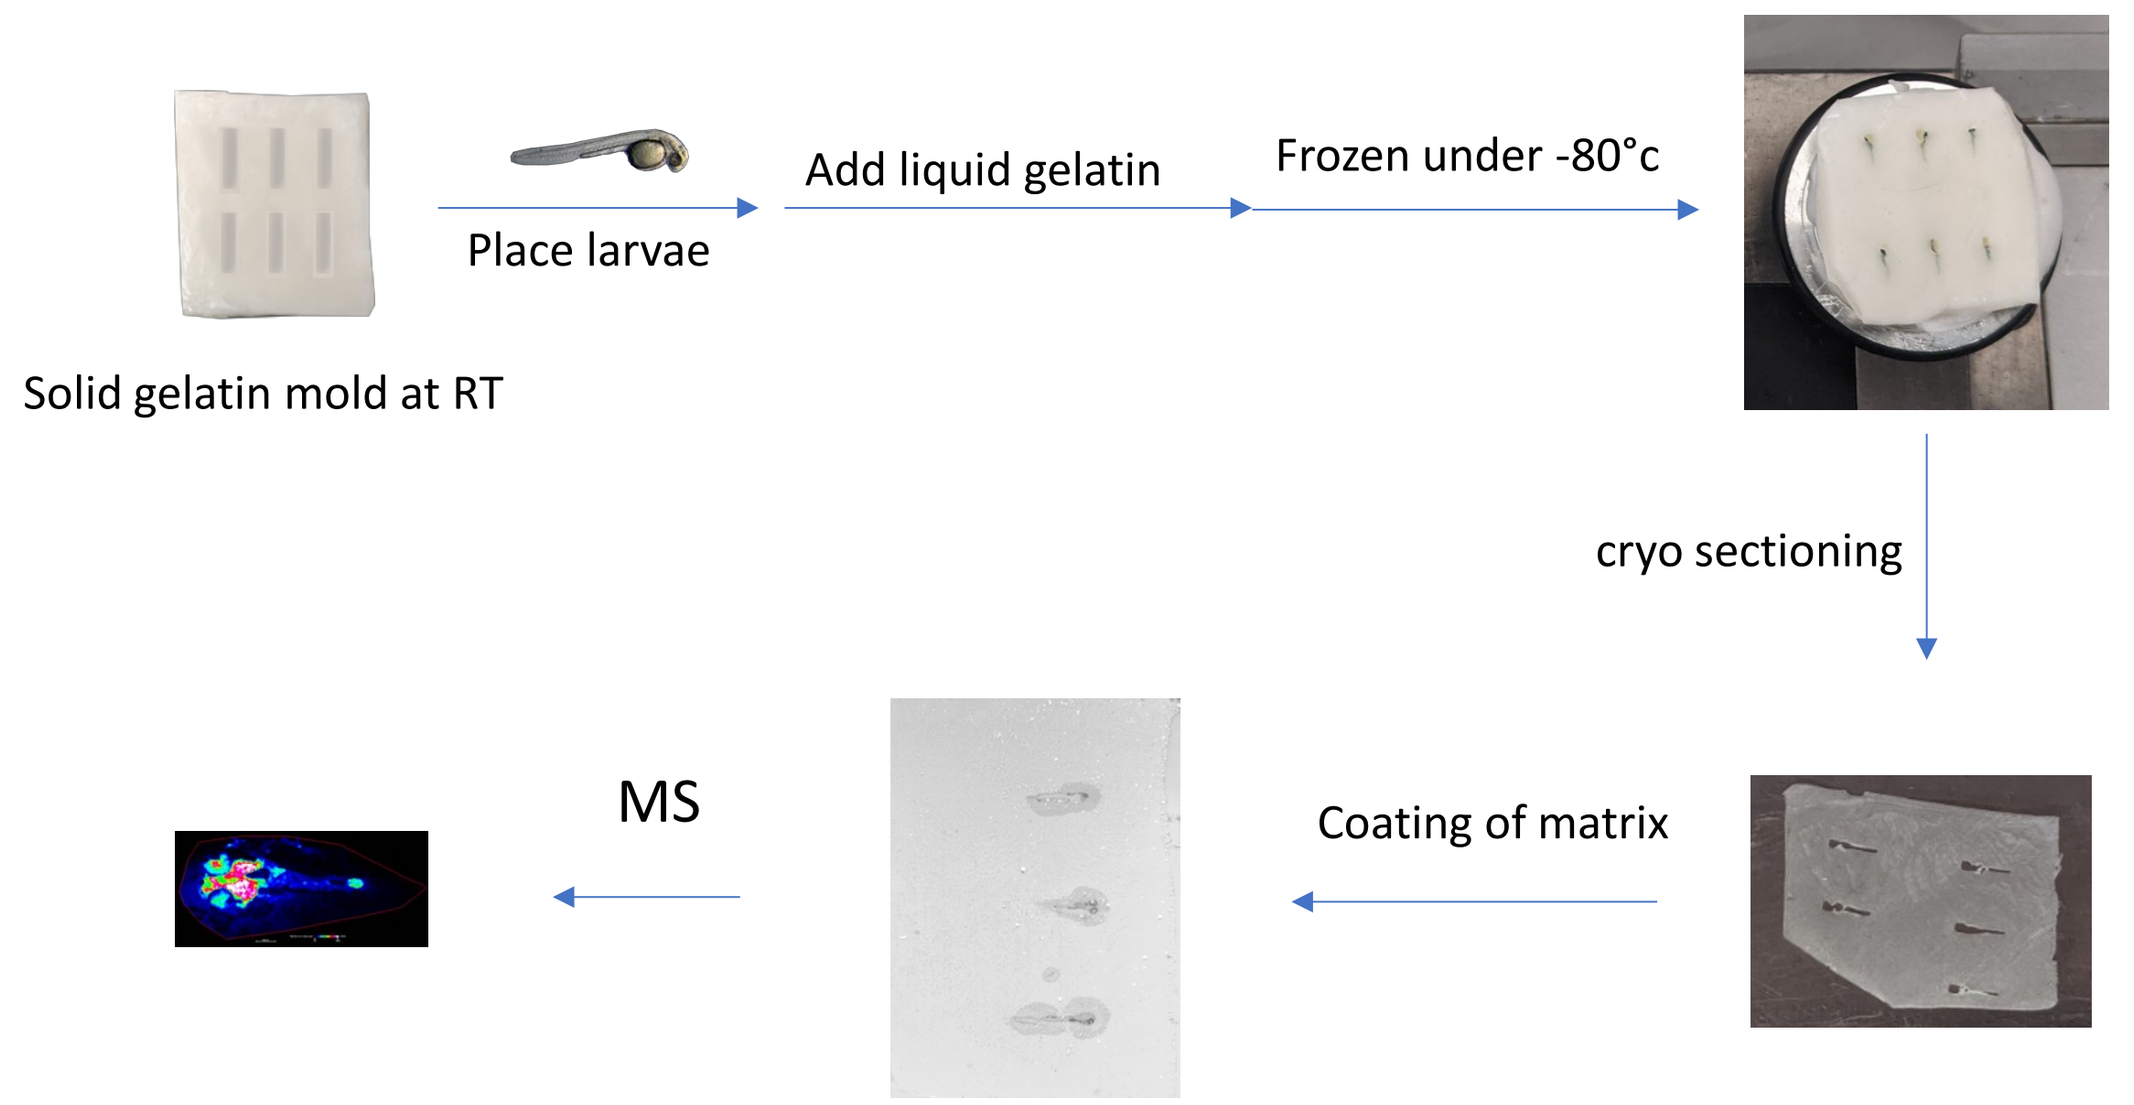
Place larvae
Add liquid gelatin
Frozen under -80°c
Solid gelatin mold at RT
cryo sectioning
MS
Coating of matrix

DESI-MS imaging of zebrafish larvae workflow

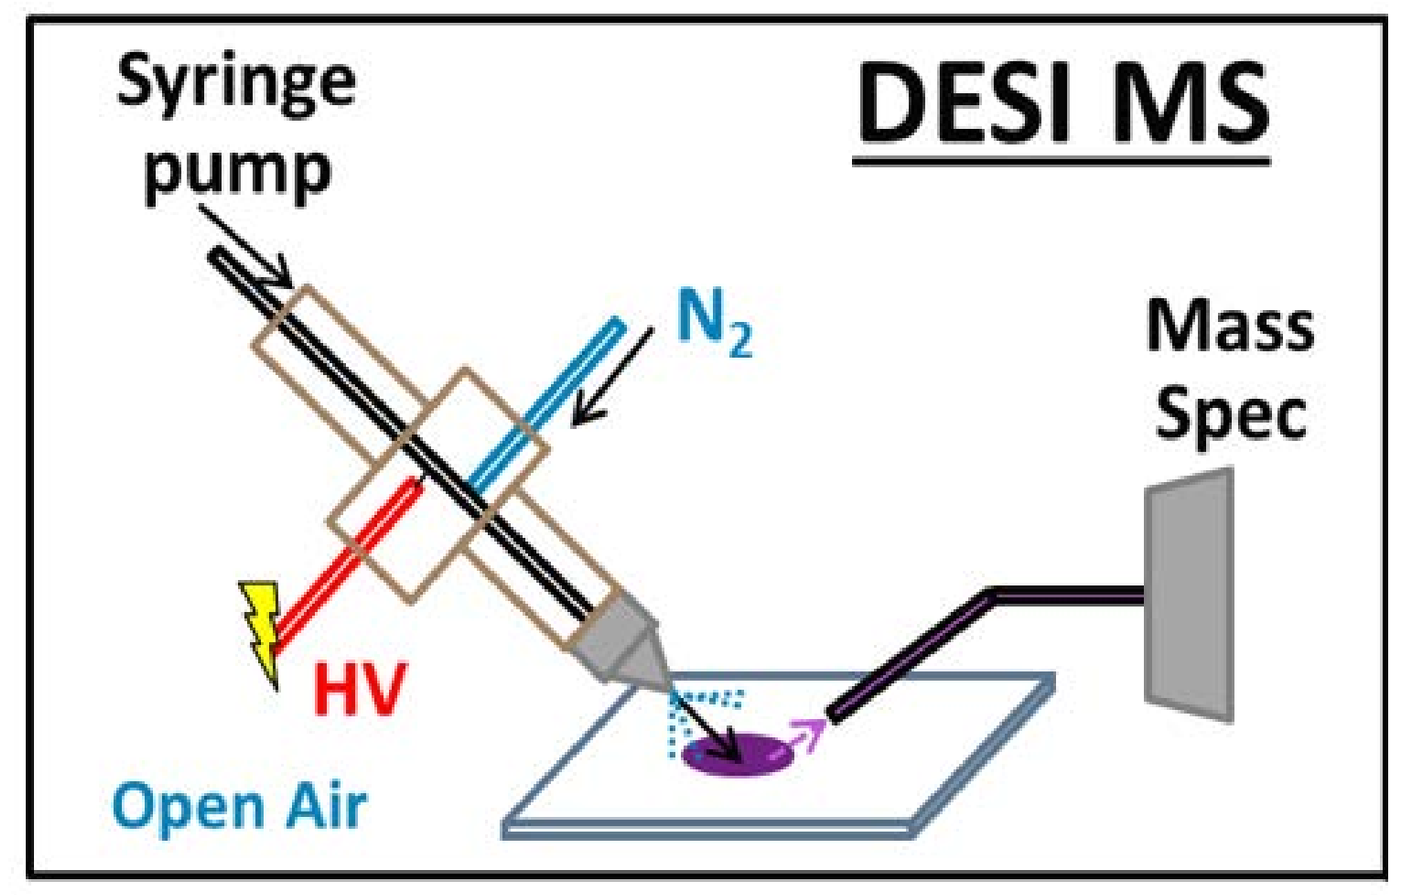
Syringe pump
DESI MS
N_2
Mass Spec
HV
Open Air

- Charged Solvent performs extraction/desorption of analyte
- Common charge and nebulizing gas remove solvent
- Simple thaw mount of tissues and analysis in ambient conditions

Control of fish placement

Better reproducibility reached with gelatin block that has fish shape surface made from 3D printed mold

Embedding Without mold

Embedding With 3D mold

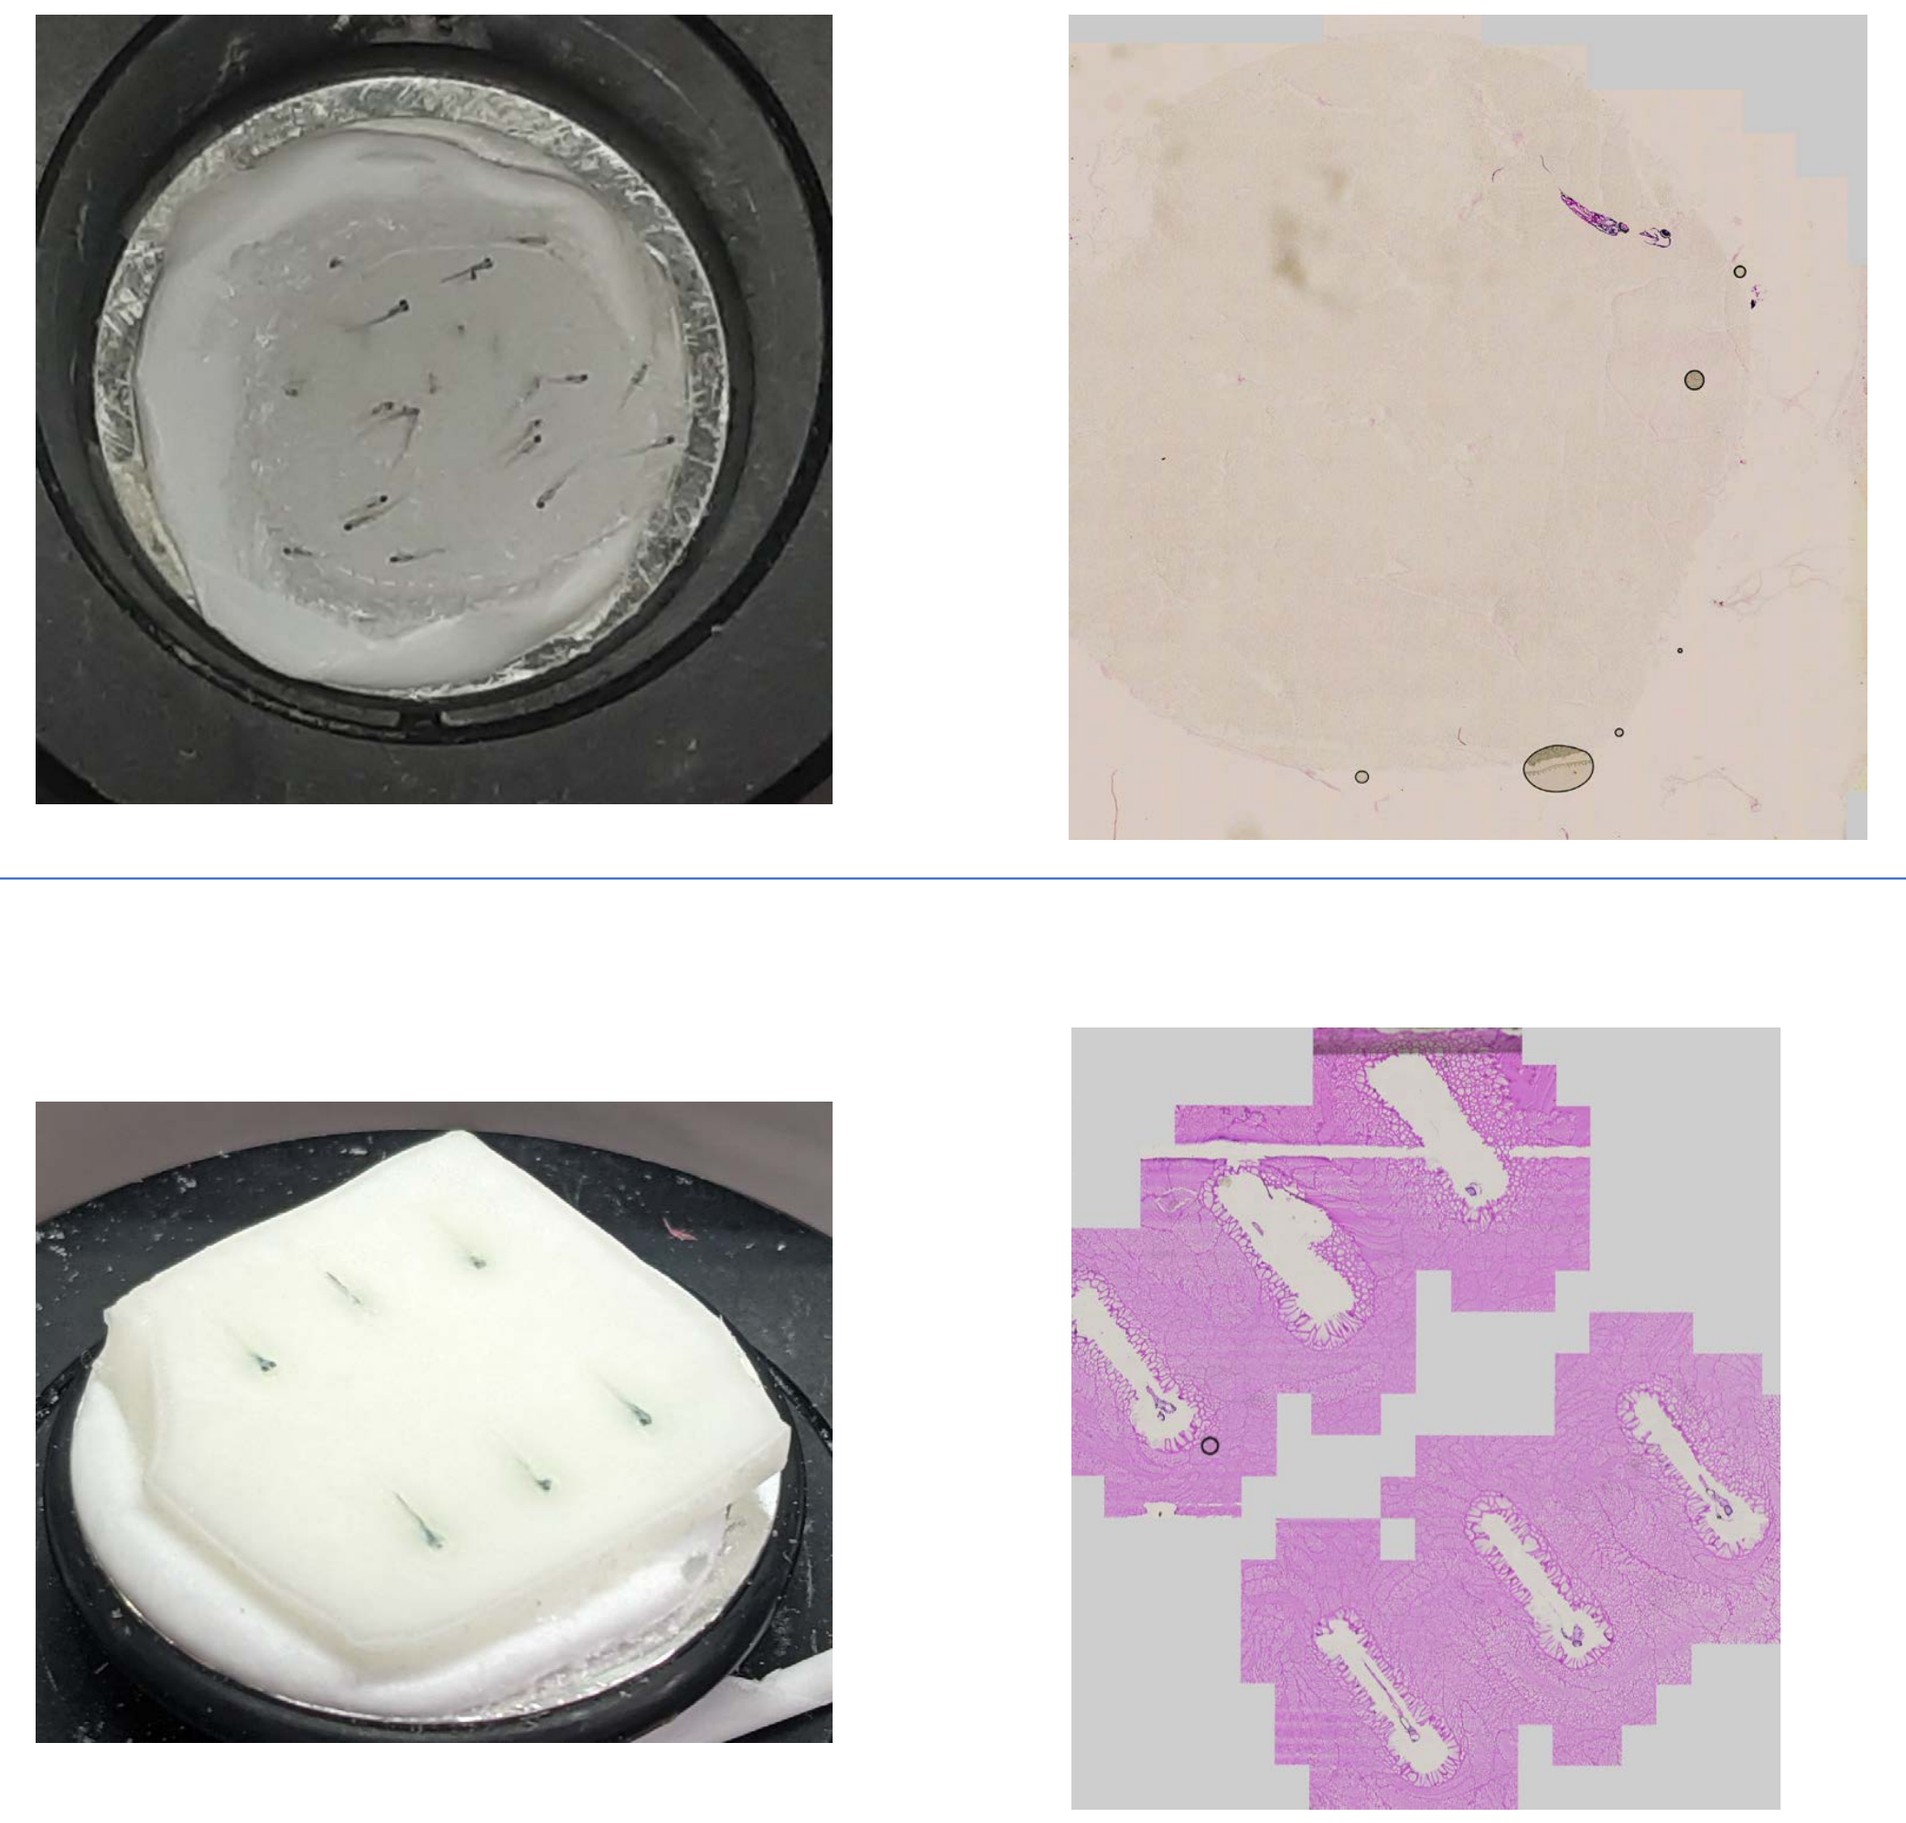

Optimization of embedding media

tested media: CMC/gelatin[1], gelatin[1], HPMC/PVP[2], pHPMA[3]
prefered media are: room temperature gelatin block (20%gelatin made at 50°C) combines liquid gelatin (20% made at 85°C)

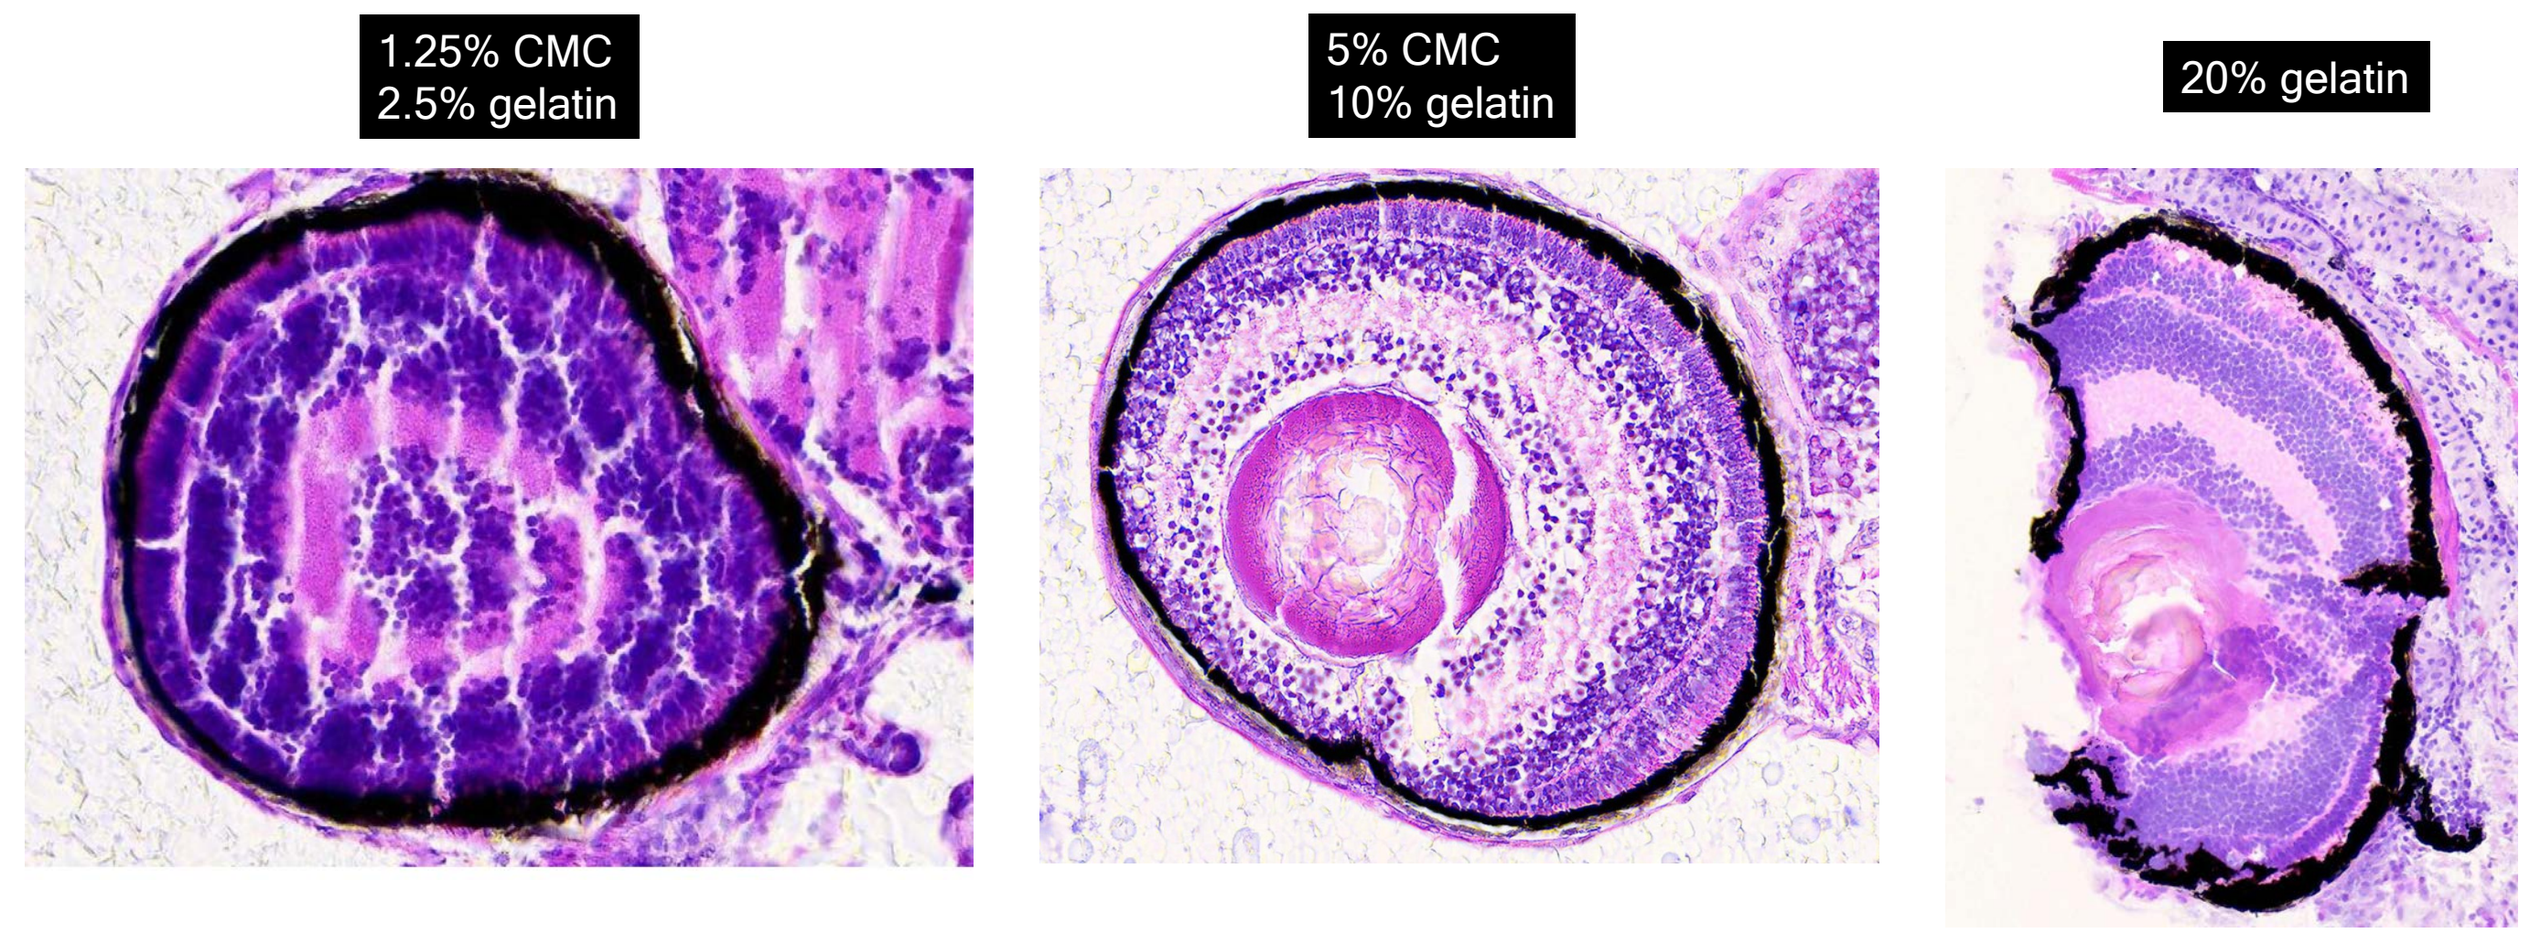
1.25% CMC
2.5% gelatin
5% CMC
10% gelatin
20% gelatin

Challenges of sectioning zebrafish larvae

with or without tape transfer

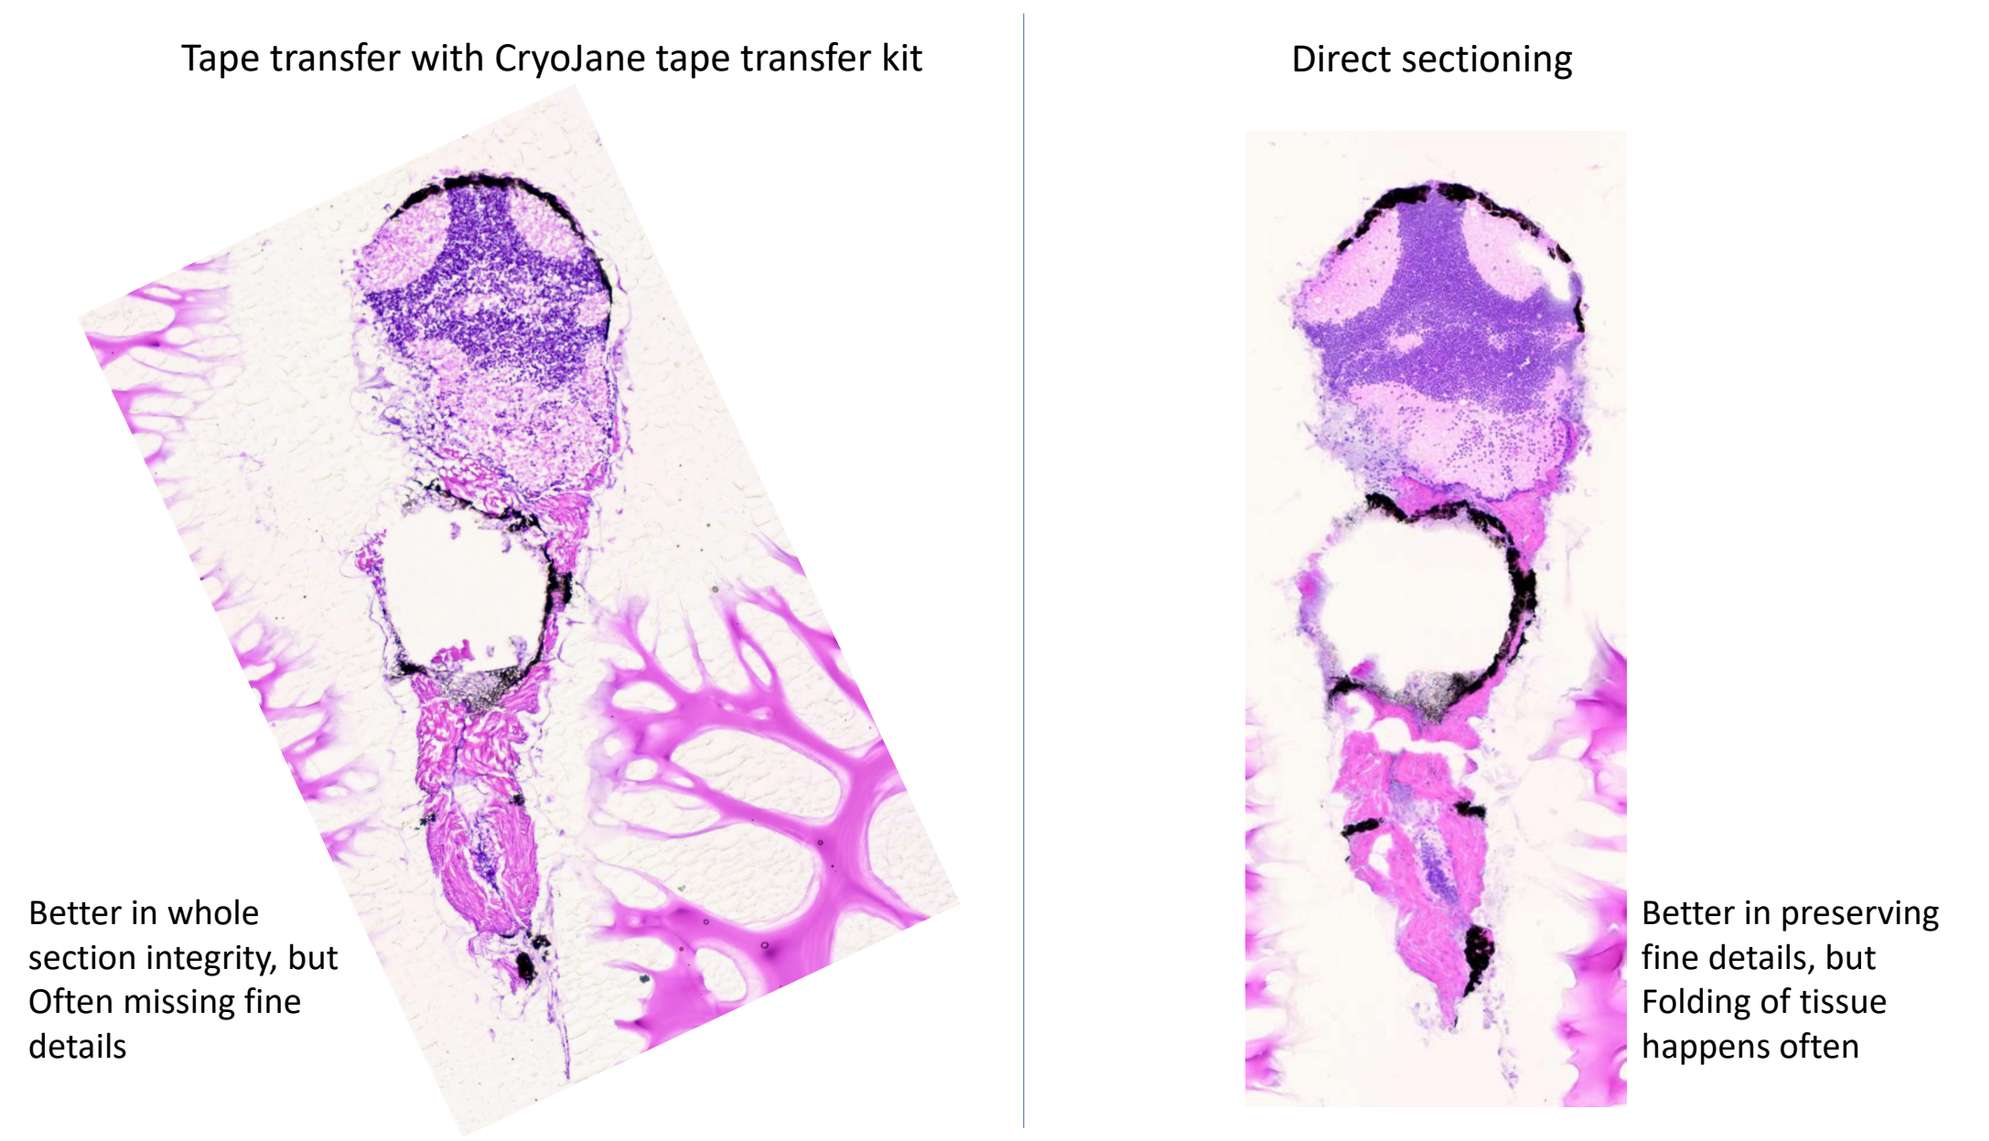
Tape transfer with CryoJane tape transfer kit
Direct sectioning
Better in whole section integrity, but Often missing fine details
Better in preserving fine details, but Folding of tissue happens often

Drug distribution in dosed zebrafish larvae

- Mass spectrometry imaging allows virtual dissection of the zebrafish larva to compare tissue specific drug distribution between molecules
- Images demonstrate greater abundance of buspirone and clozapine in the CNS relative to loperamide in accord with mammalian tissue distribution

MALDI imaging results

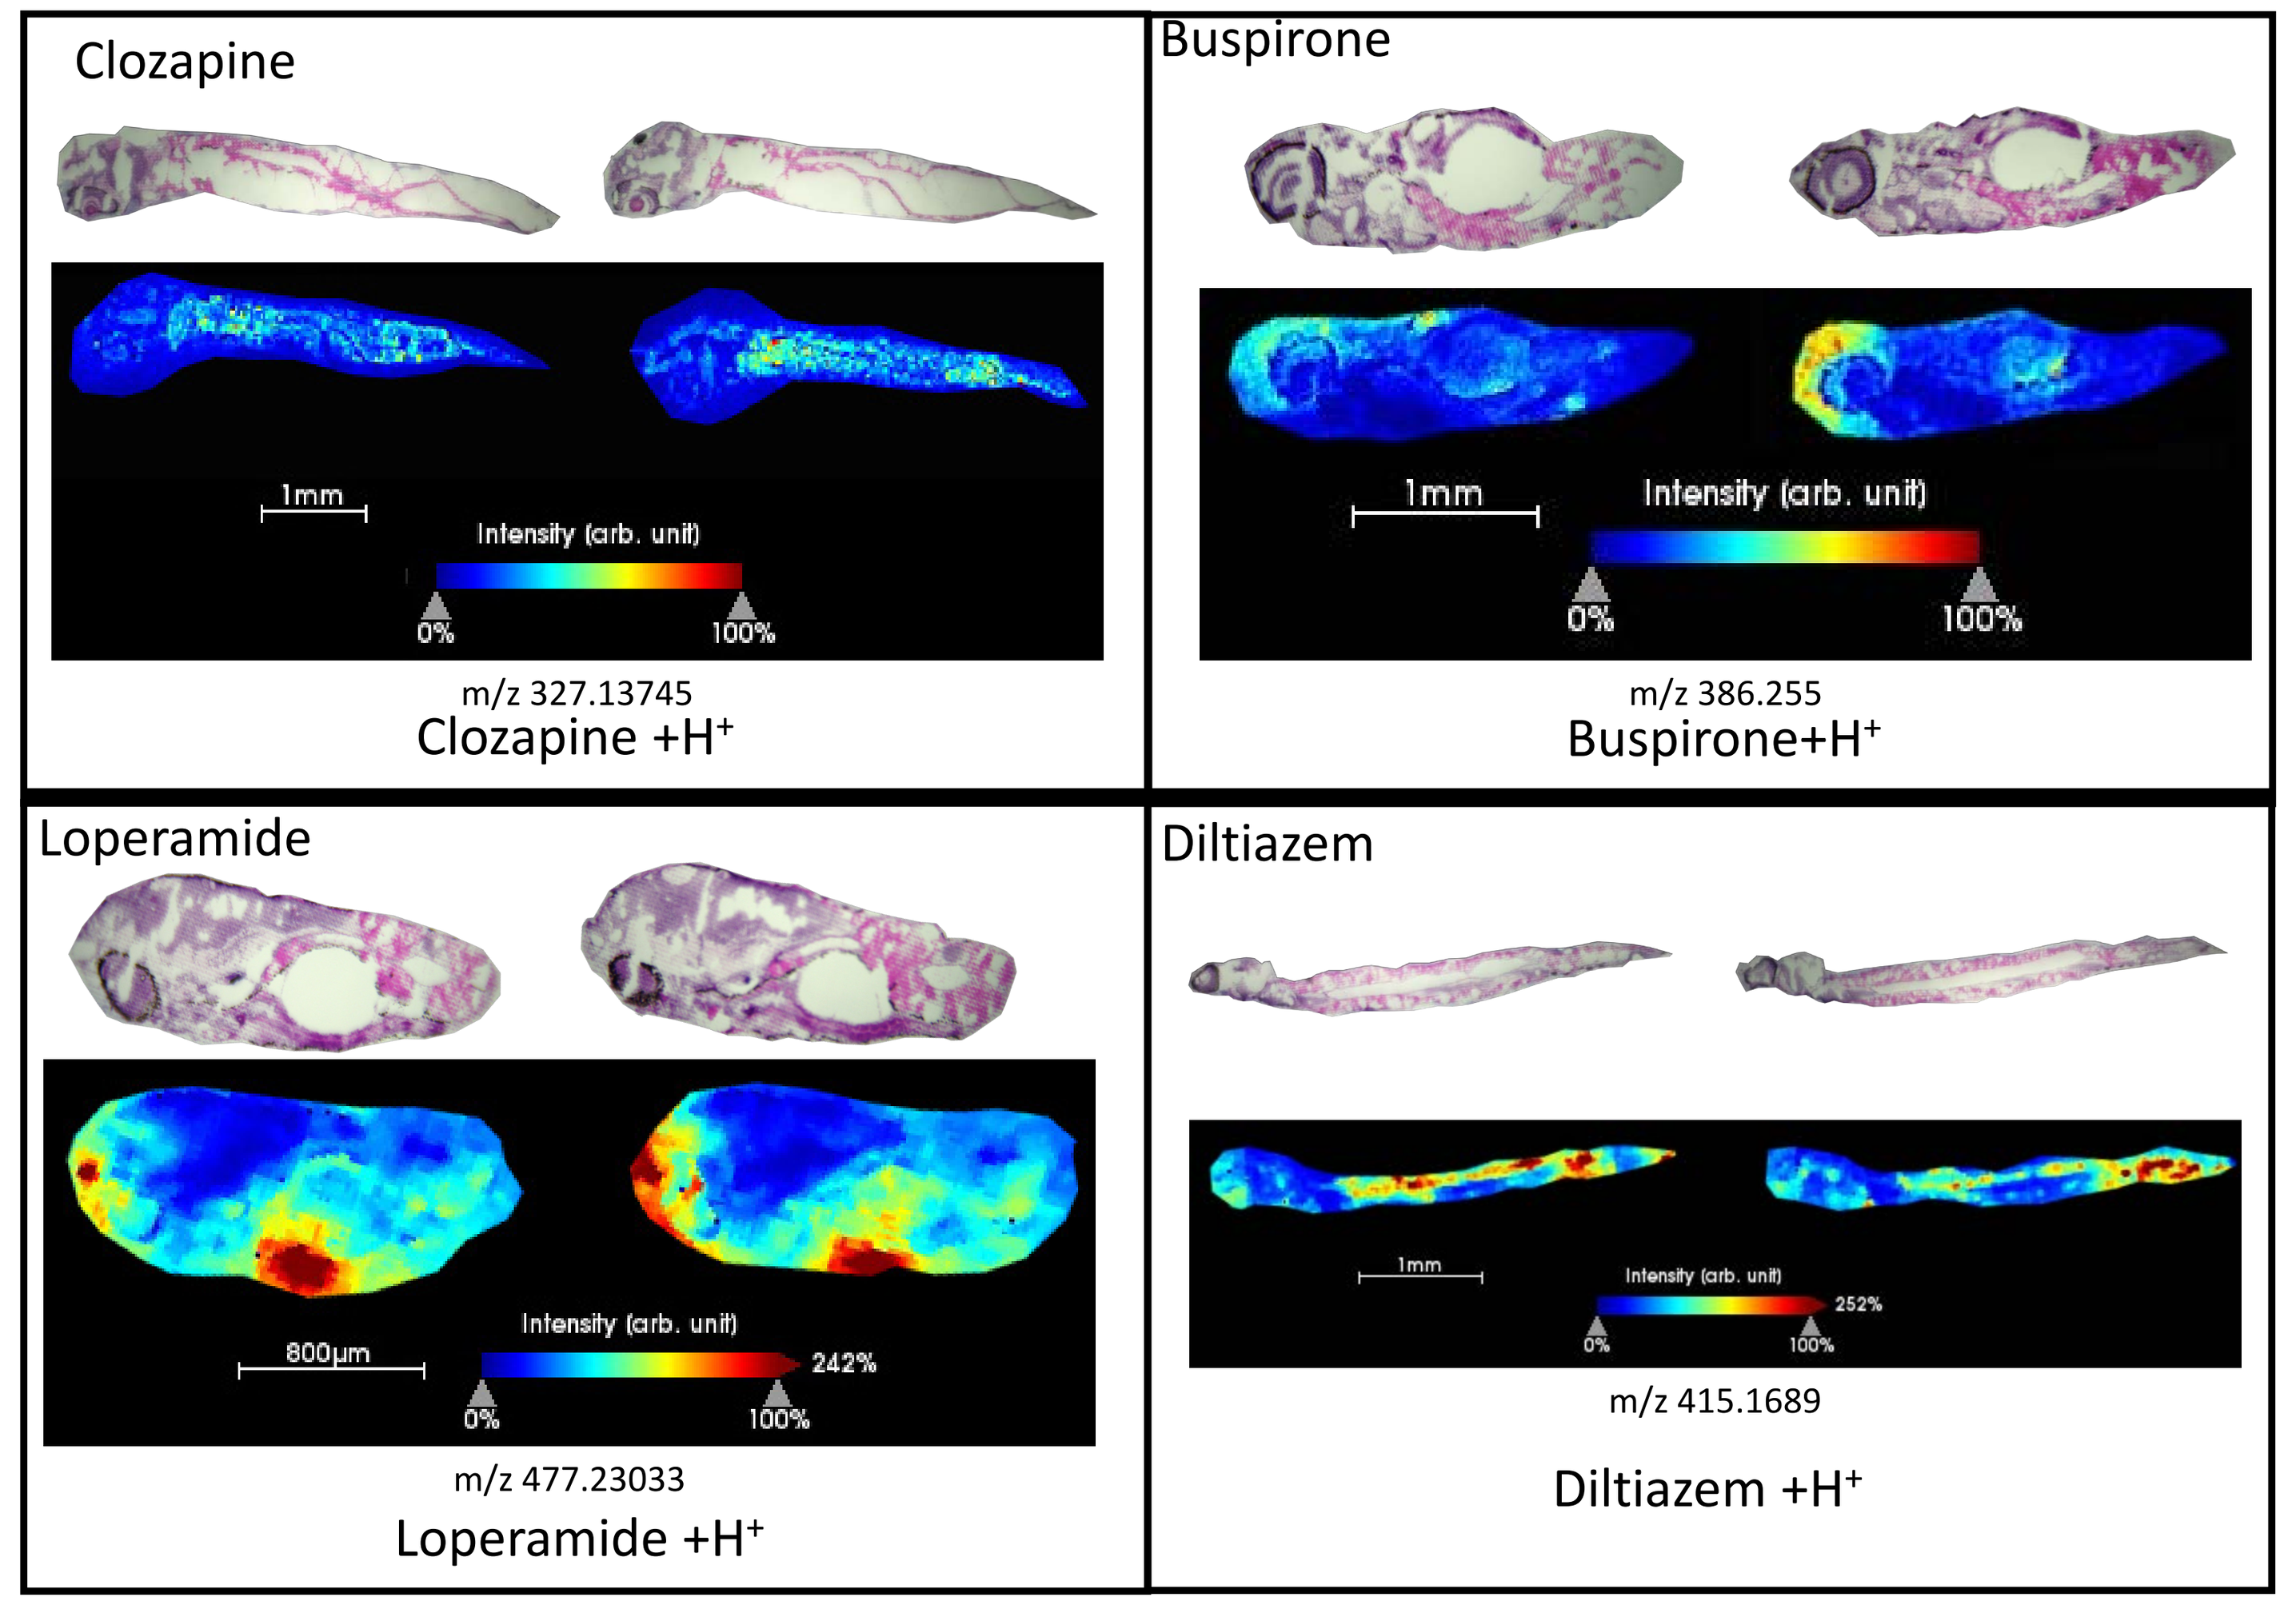
Clozapine
1mm
Intensity (arb. unit)
0%
100%
m/z 327.13745
Clozapine +H+
Buspirone
1mm
Intensity (arb. unit)
0%
100%
m/z 386.255
Buspirone+H+
Loperamide
800µm
Intensity (arb. unit)
242%
0%
100%
m/z 477.23033
Loperamide +H+
Diltiazem
1mm
Intensity (arb. unit)
252%
0%
100%
m/z 415.1689
Diltiazem +H+

DESI imaging results

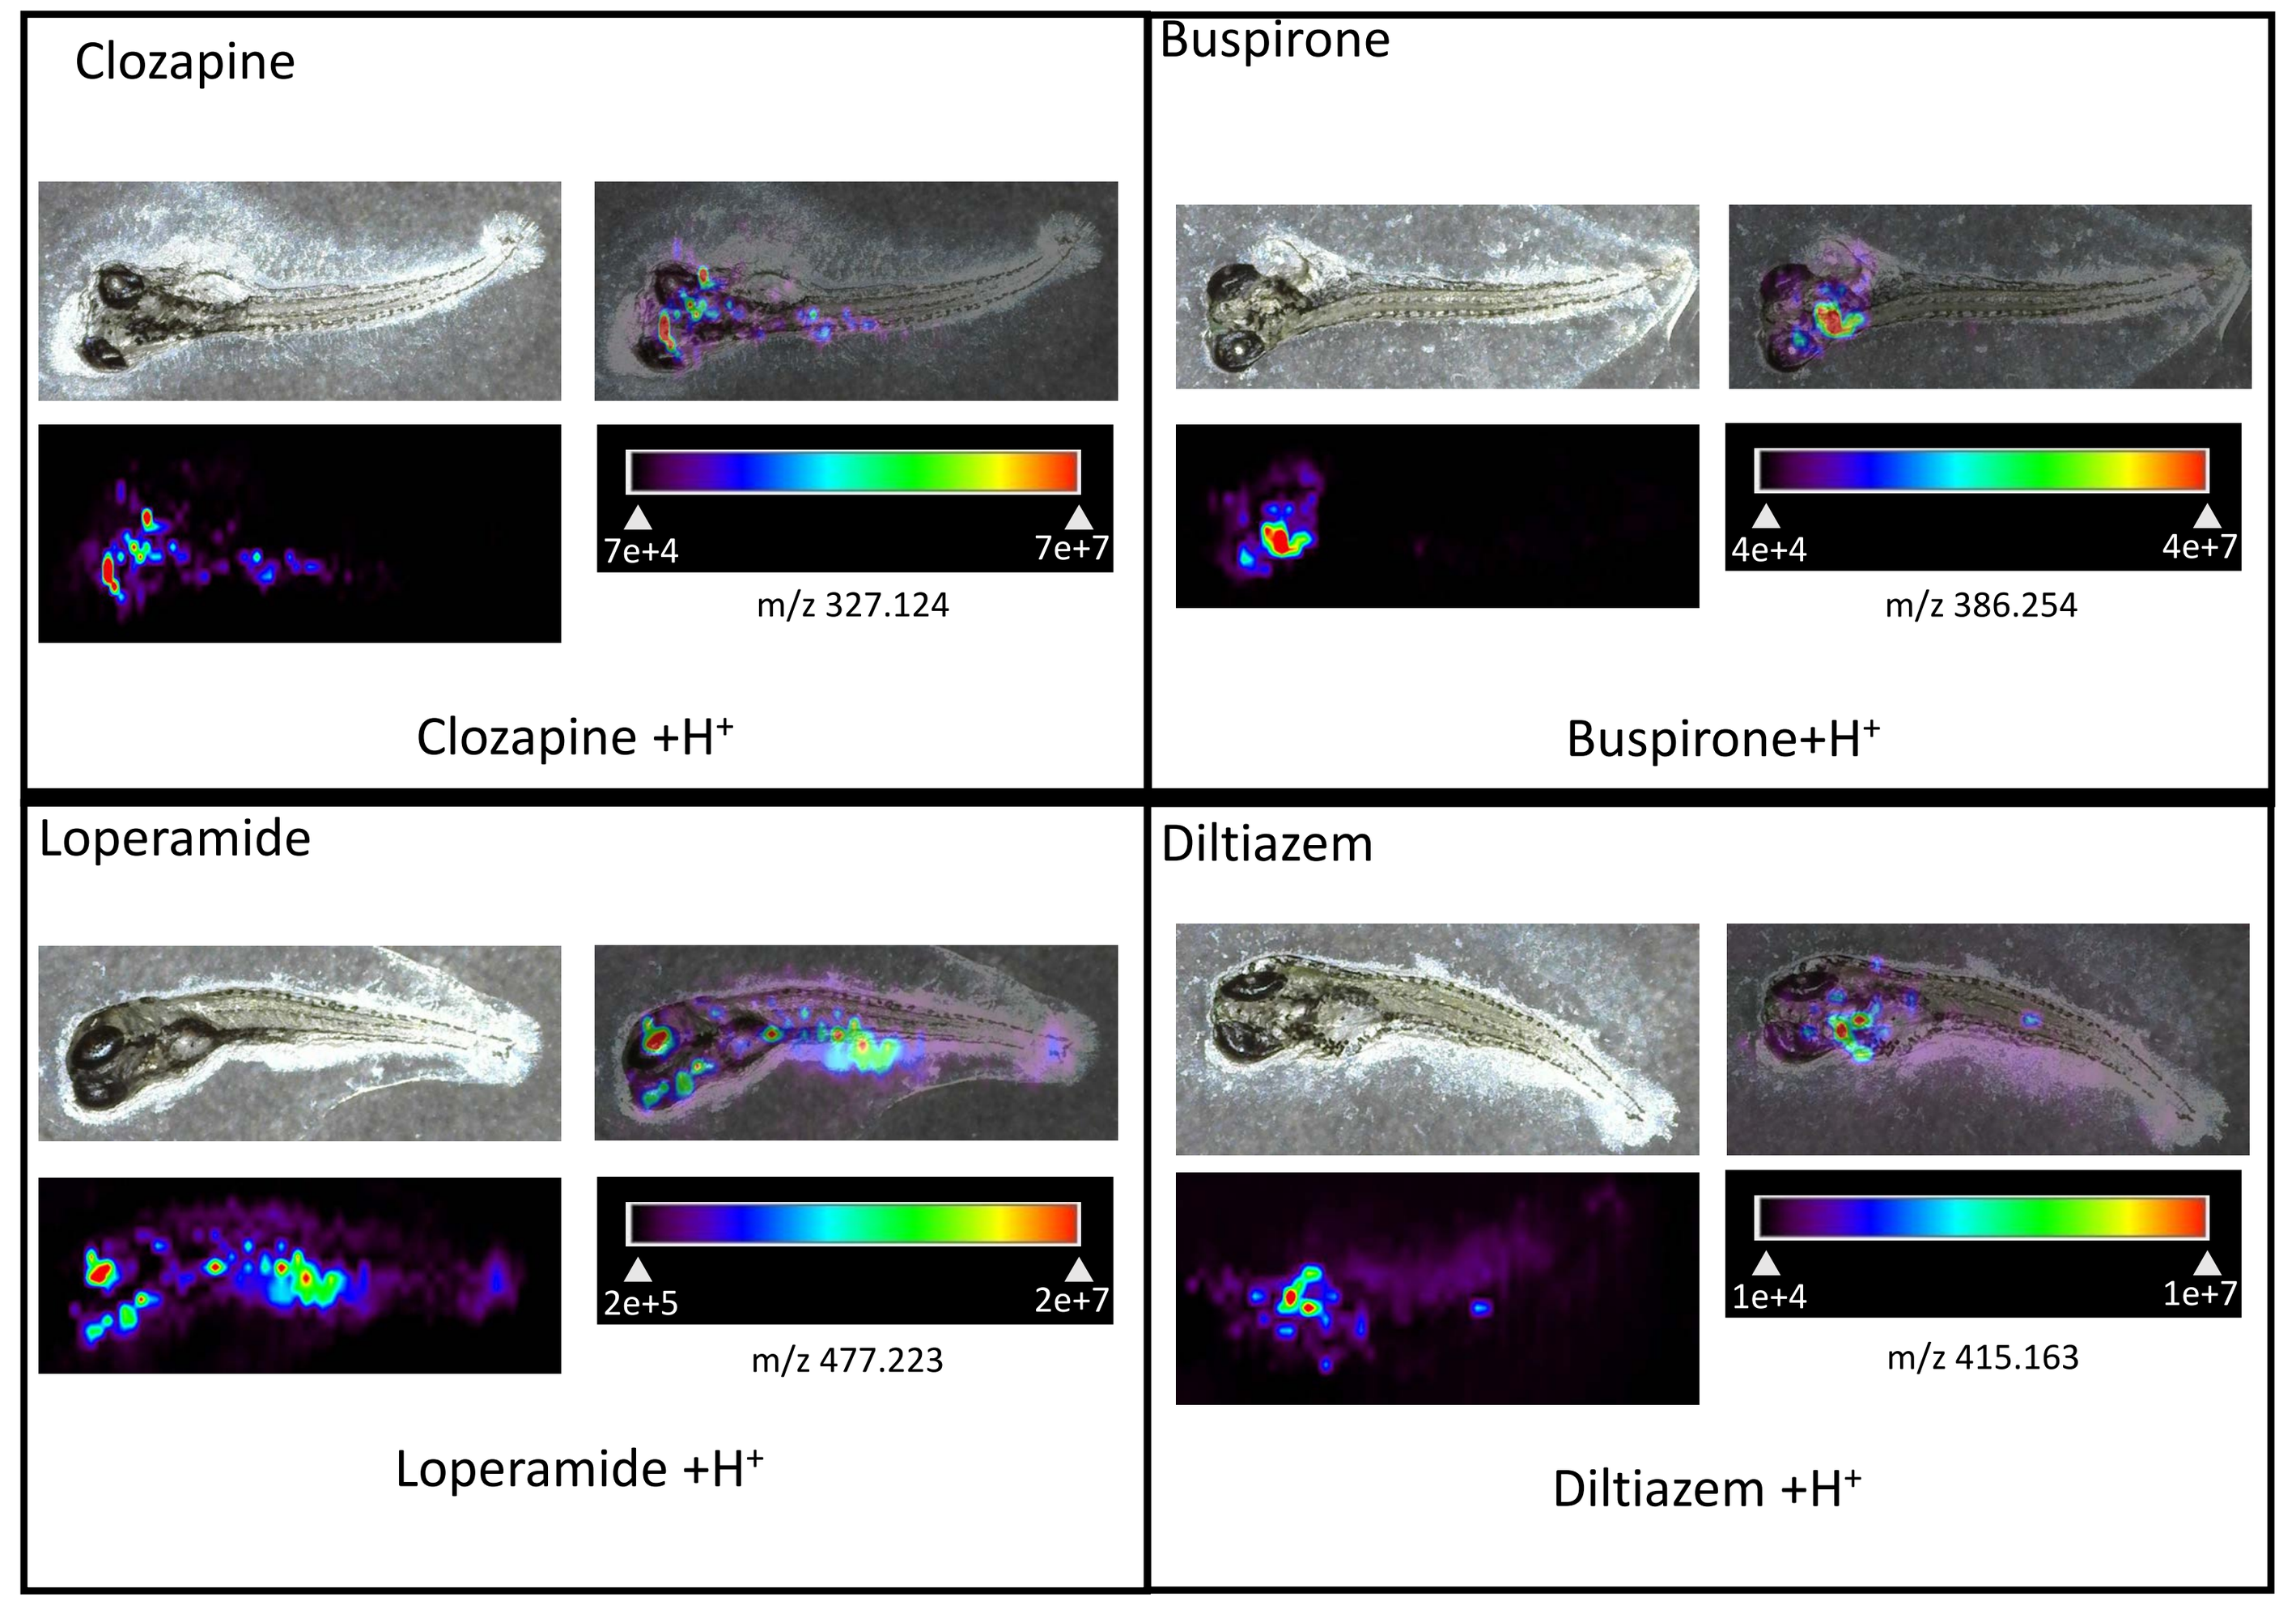
Clozapine
7e+4
7e+7
m/z 327.124
Clozapine +H+
Buspirone
4e+4
4e+7
m/z 386.254
Buspirone+H+
Loperamide
2e+5
2e+7
m/z 477.223
Loperamide +H+
Diltiazem
1e+4
1e+7
m/z 415.163
Diltiazem +H+

Zebrafish organ ion marker by MALDI

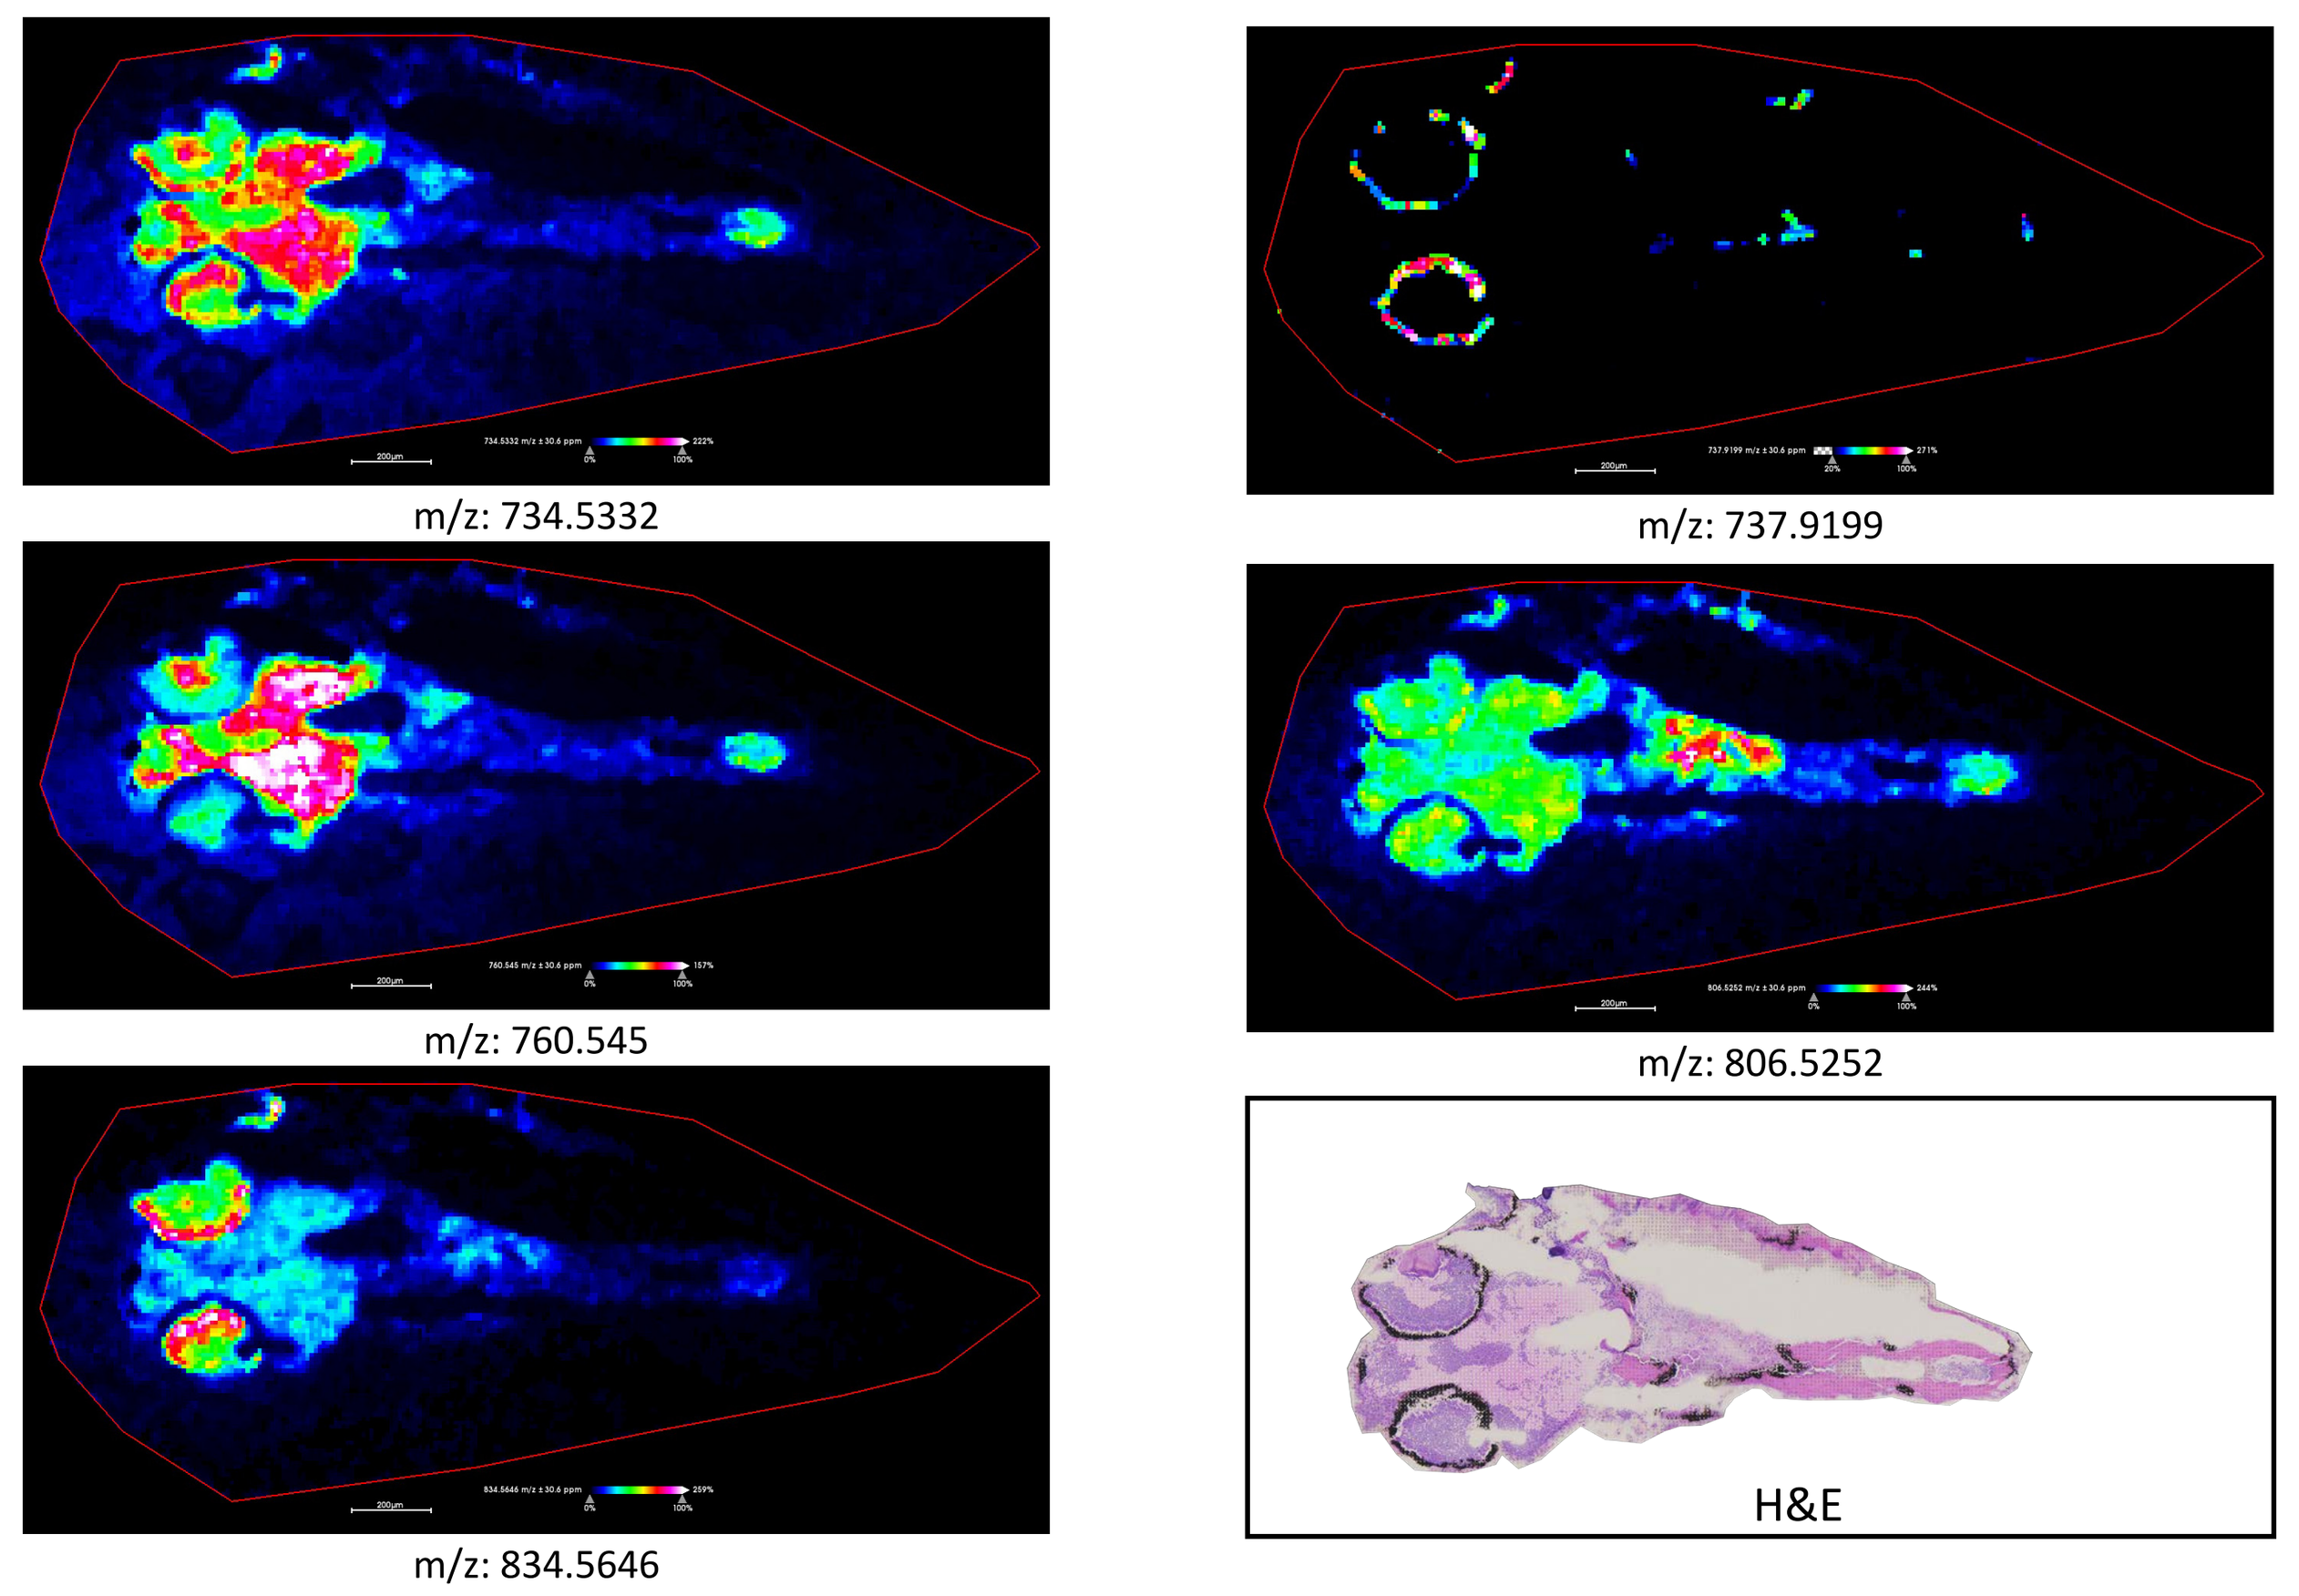
m/z: 734.5332
m/z: 737.9199
m/z: 760.545
m/z: 806.5252
m/z: 834.5646
H&E

Summary

- Zebrafish larvae embedding media and method were optimized.
- Gelatin made at 50°C stays solid at RT and gelatin made at 85°C stays liquid at RT. Ambient temperature placement of fish larvae makes the operation feasible to control the fish body orientation for obtaining of quality sections.
- Zebrafish larvae as toxicity model was demonstrated. The distribution of model compounds correlate the organ ion marker well, indicating the usefullness of using zebrafish larvae as funneling tool in early drug developement.
- Further imaging of zebrafish larvae to determine more organ ion marker is under process to facilitate the correlation between drug distribution and organs.
- Comparing MALDI to DESI, MALDI imaging provides higher spatial resolution, DESI has minmal sample preparation.

References:

1) J Biomol Tech. 2013, Sep,24(3):119-27
2) Anal. Chem. 2020, 92, 16, 11080-11088
3) Anal. Chem. 2011, 83, 5458-5462

Disclosures:

- All authors are employees of AbbVie and may own AbbVie stock.
- AbbVie sponsored and funded the study; contributed to the design; participated in the collection, analysis, and interpretation of data, and in writing, reviewing, and approval of the final presentation.